BOLD-100-001 (TRIO039): A Phase 1b/2a Study of BOLD-100 in Combination with FOLFOX Chemotherapy in Patients with Pre-Treated Advanced Gastric and Biliary Tract Cancer: Efficacy and Safety Analysis

Abstract #4098

Grainne M. O'Kane, MD[1]; Jennifer L. Spratlin, MD[2]; Do-Youn Oh, MD[3]; Sun Young Rha, MD[4]; Elena Elimova, MD[5]; Petr Kavan, MD[6]; Moon Ki Choi, MD[7]; Rachel Anne Goodwin, MD[8]; Seung Tae Kim, MD[9]; Dong-Hoe Koo, MD[10]; Khalif Halani[11]; E Russell McAllister[12]; Michelle Jones[12]; Malcolm Snow[12]; Yasmin Lemmerick[13]; Gonzalo Spera[14]; Jim Pankovich[12]

[1]Trinity St. James Cancer Institute, Dublin, Ireland; [2]Cross Cancer Institute, University of Alberta, Edmonton, Canada; [3]Seoul National University Hospital, Seoul, Korea, Republic of (South); [4]Yonsei Cancer Center, Yonsei Cancer Center, Yonsei University Health System, Seoul, South Korea; [5]Princess Margaret Cancer Centre, University Health Network, Toronto, Canada; [6]Sir Mortimer B. Davis Jewish General Hospital, Montreal, Canada; [7]National Cancer Center, Goyang-Si, South Korea; [8]The Ottawa Hospital Research Institute, Ottawa, Canada; [9]Samsung Medical Center, Seoul, South Korea; [10]Kangbuk Samsung Hospital, Seoul, South Korea; [11]EMMES, Burnaby, Canada; [12]Bold Therapeutics, Inc., Vancouver, Canada; [13]Translational Research in Oncology, Edmonton, Canada; [14]Translational Research in Oncology, Montevideo, Uruguay

Introduction

- BOLD-100 is a first in class ruthenium-based anticancer agent in development for the treatment of gastrointestinal (GI) cancers.
- BOLD-100 is currently being tested in a Phase 2 clinical trial in combination with standard of care FOLFOX in patients with advanced GI cancers (NCT04421820) and has potential in a range of solid and liquid cancer indications.[1]
- BOLD-100 exerts its function via the modulation of the unfolded protein response via GRP78 downregulation, with secondary mechanistic pathways including generation of reactive oxygen species, DNA damage, modulation of lipid metabolism, and interactions with ribosomal proteins.
- Here, we present interim efficacy, safety and tolerability data in patients with heavily pre-treated advanced gastric (GC) and biliary tract cancers (BTC).

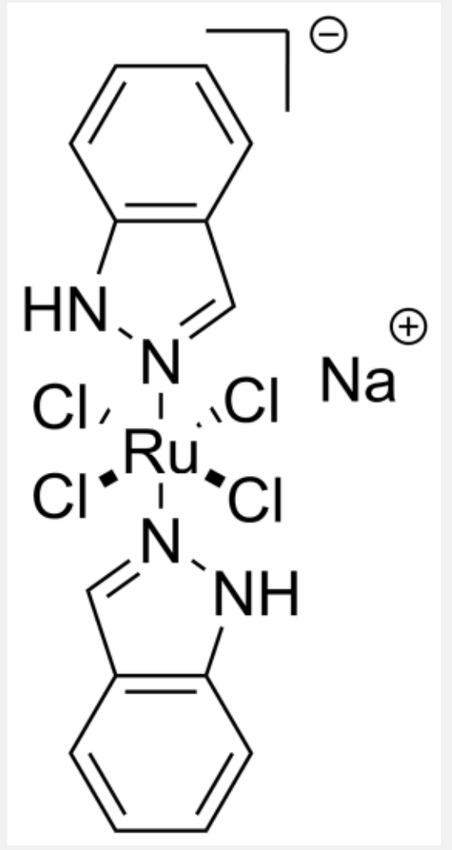
HN
N
Cl
Cl
Ru
Cl
Cl
N
NH
Na

BOLD-100

Methods

Figure 1. Study Design

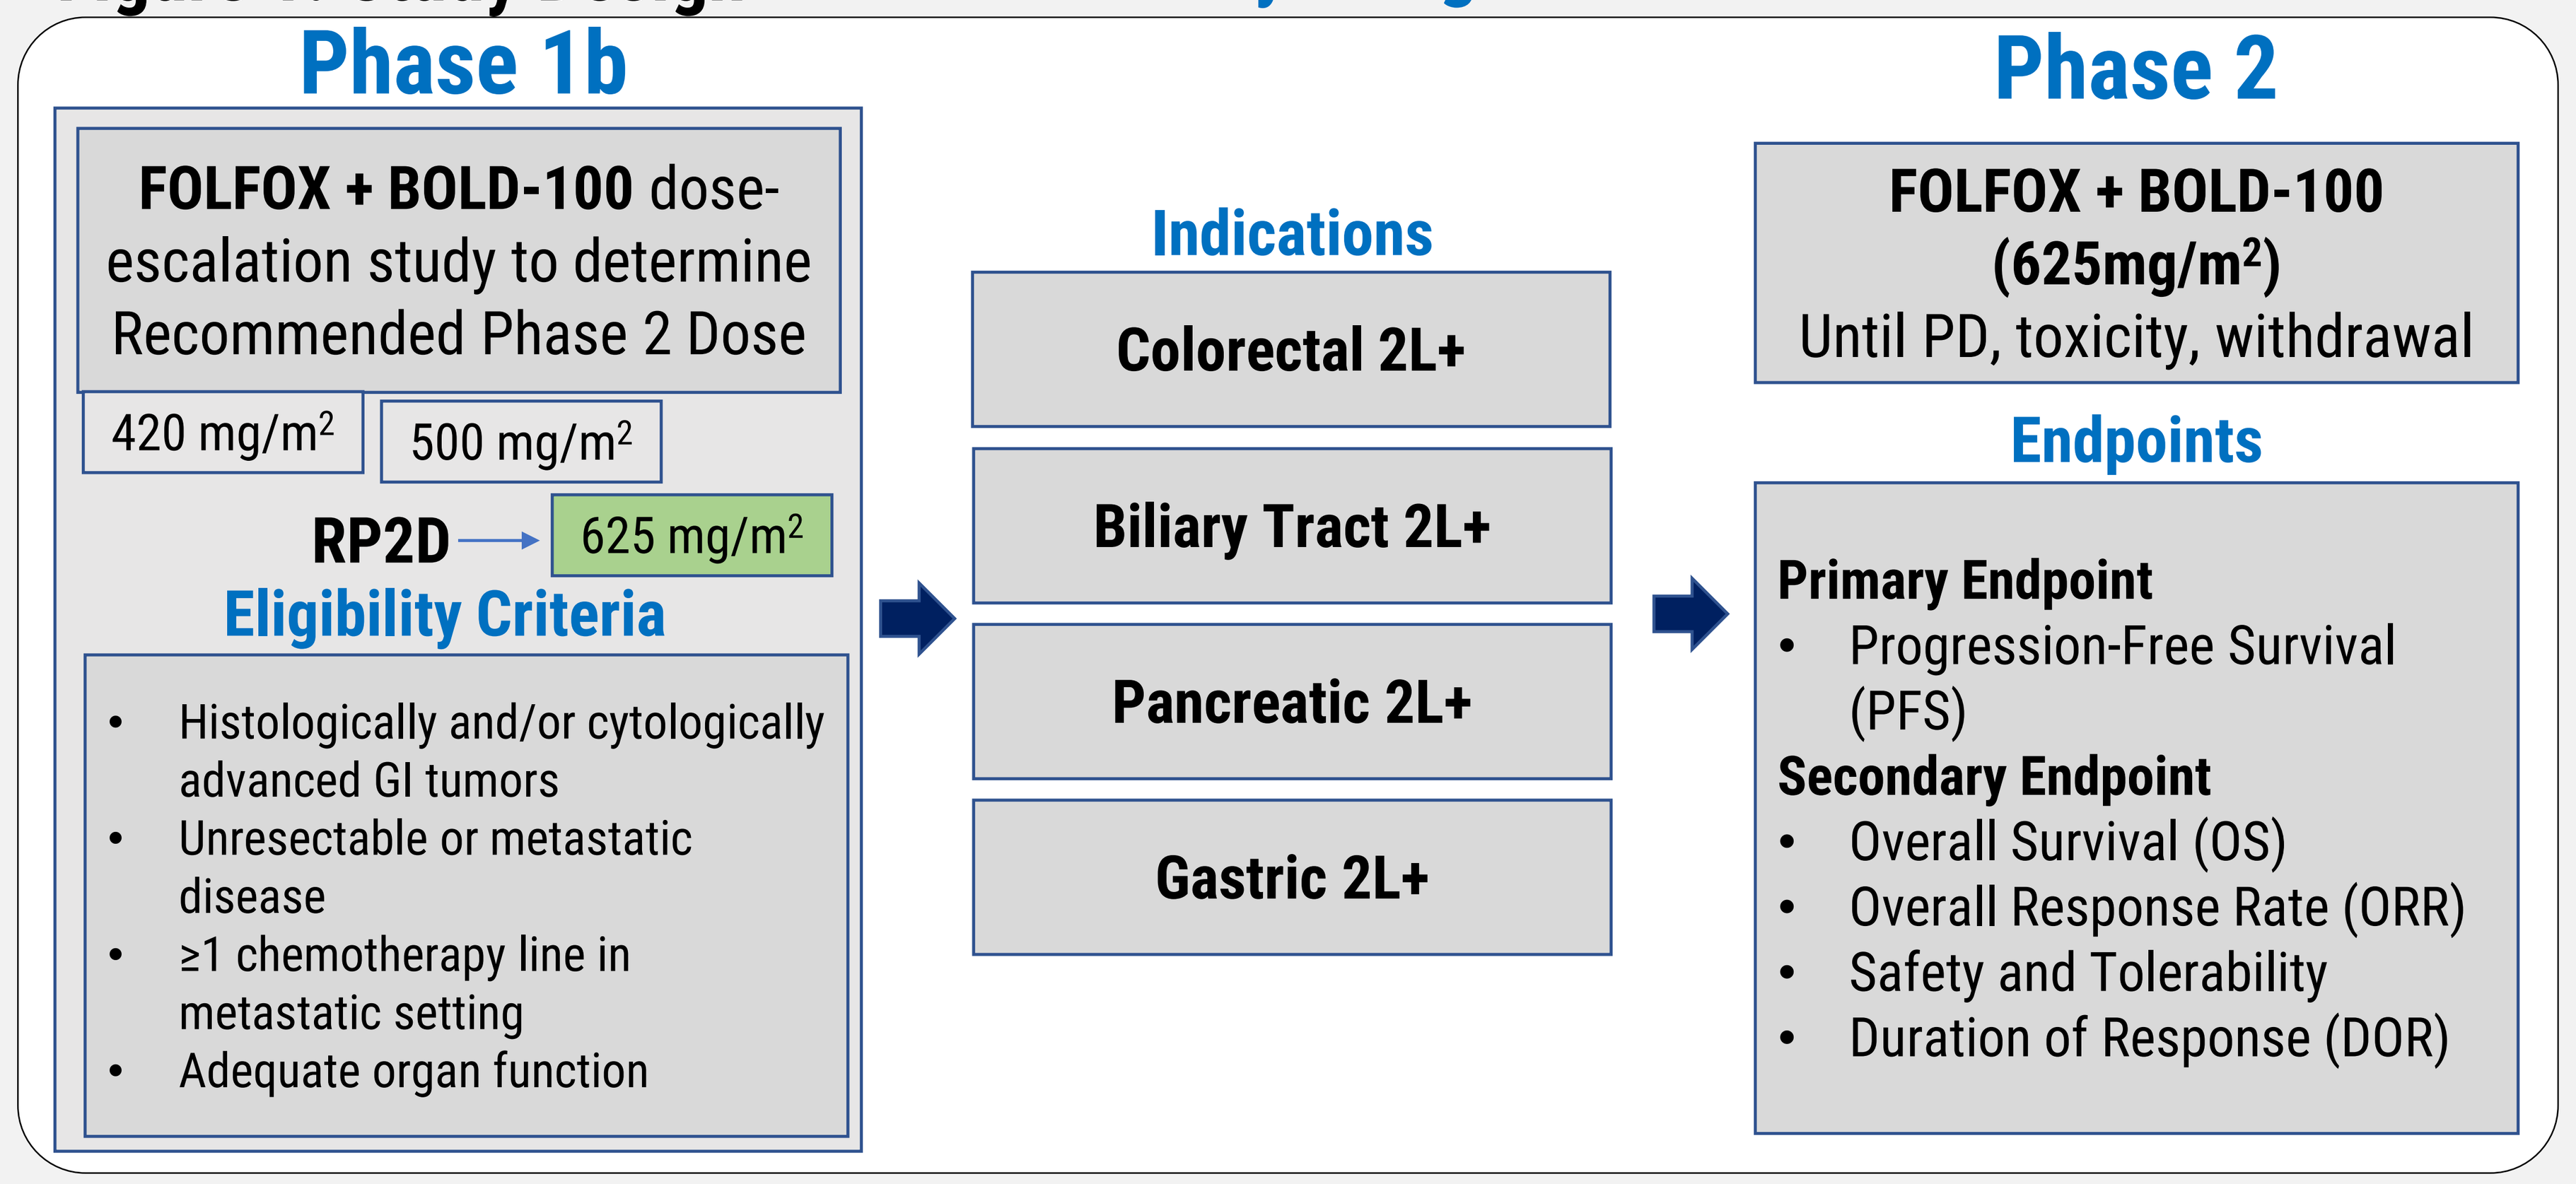
Study Design
Phase 1b
FOLFOX + BOLD-100 dose-escalation study to determine Recommended Phase 2 Dose
420 mg/m2
500 mg/m2
RP2D
625 mg/m2
Eligibility Criteria
Histologically and/or cytologically advanced GI tumors
Unresectable or metastatic disease
≥1 chemotherapy line in metastatic setting
Adequate organ function
Indications
Colorectal 2L+
Biliary Tract 2L+
Pancreatic 2L+
Gastric 2L+
Phase 2
FOLFOX + BOLD-100 (625mg/m2)
Until PD, toxicity, withdrawal
Endpoints
Primary Endpoint
Progression-Free Survival (PFS)
Secondary Endpoint
Overall Survival (OS)
Overall Response Rate (ORR)
Safety and Tolerability
Duration of Response (DOR)

FOLFOX regimen: oxaliplatin 85 mg/m^2 IV Q2W; leucovorin 400 mg/m^2 IV Q2W; and 5-FU 2400 mg/m^2 (continuous 46 hour infusion). 5-FU, 5-fluorouracil; IV, intravenously; Q2W, once every 2 weeks; RP2D, Recommended phase 2 dose; 2L+. Second line and beyond.

Statistical Analysis

- Safety analyses included all patients who received ≥1 dose of any study drug
- Efficacy analyses included all patients who had a baseline and ≥1 post-baseline assessment or discontinued study treatment due to progressive disease or death
 - Clinical activity was assessed via RECIST v1.1 criteria
 - Disease control rate (DCR) was defined as the percentage of patients with a best overall response of complete response (CR), partial response (PR), or stable disease (SD)
 - A Bayesian statistical approach was used in this study.

Results

- As of Dec 31, 2022, 35 patients with stage IV disease were enrolled and treated in these study arms **(Table 1)**.
 - 13 patients were treated, 9 patients were evaluable for efficacy in the GC arm
 - 22 patients were treated, 18 patients were evaluable for efficacy in the BTC arm

Gastric Cancer

- Three (3) median prior therapies
- 10/13 patients had prior platinum based treatment
- 5 patients (38%) remain on treatment and six (46%) remain in follow-up

Biliary Tract Cancer

- Two (2) median prior therapies
- 21/22 patients had prior Gemcitabine + Cisplatin treatment
- 4 patients (18%) remain on treatment and three (14%) remain in follow-up

Table 1. Demographics and Disease Characteristics

	Gastric Cancer (N = 13)	Biliary Tract Cancer (N = 22)
Median age (range), yrs	61 (49–84)	61 (33-81)
Male sex, n (%)	9 (69)	12 (55)
Race		
White	0 (0)	8 (36)
Asian	13 (100)	14 (64)
ECOG Performance		
0	5 (38)	10 (45)
1	8 (62)	12 (55)
Stage IV disease	13 (100)	22 (100)
Median prior therapies	3 (0-7)	2 (1-5)
Prior Platinum, n (%)	10 (77)	-
Prior GemCis, n (%)	-	21 (95)

GemCis refers to first line (1L) Gemcitabine and Cisplatin

Table 2. Biliary Tract Cancer Primary Tumour Site, n (%)

Distal	8 (36)
Gallbladder	5 (23)
Intrahepatic	5 (23)
Perihilar	1 (5)
Unknown	3 (14)

SAFETY

Table 3 summarizes the TEAEs related to BOLD-100 + FOLFOX in each indication. For all treated patients (n = 35), 33 reported 1 or more TEAE; the most common TEAEs were neutrophil count decrease (n = 15, 43%), nausea (n = 10, 29%), fatigue (n = 7, 20%), and anaemia (n = 5, 14%).

- Most TEAEs were Grade 1-2. 14 patients (40%) reported Grade 3/4 neutrophil count decreased AEs

Table 3. Summary of Treatment Emergent Adverse Events (TEAEs) Related to BOLD-100 + FOLFOX ≥20%

	Gastric Cancer (N = 13)		Biliary Tract Cancer (N = 22)	
	Any Grade	Grade ≥ 3	Any Grade	Grade ≥ 3
Any TEAE[a]	10 (77)	7 (58)	21 (95)	16 (73)
Neutrophil count decreased	5 (38)	5 (38)	10 (45)	9 (41)
Nausea	2 (15)	0 (0)	8 (36)	0 (0)
Fatigue	0 (0)	0 (0)	7 (32)	0 (0)
Pyrexia	0 (0)	0 (0)	6 (27)	0 (0)
Peripheral sensory neuropathy	3 (23)	0 (0)	6 (27)	1 (4)
Platelet count decreased	3 (23)	0 (0)	5 (23)	1 (4)
Urticaria	3 (23)	0 (0)	0 (0)	0 (0)

Data are reported as number of patients, n (%). **a.** All AEs were recorded using the Medical Dictionary for Regulatory Activities (MedDRA) with severity graded by investigators according to the National Cancer Institute Common Terminology Criteria for Adverse Events (NCI CTCAE) version 5.0.

Figure 2. Change in Target Lesion Size Over Time in Response-evaluable Patients with GC and BTC

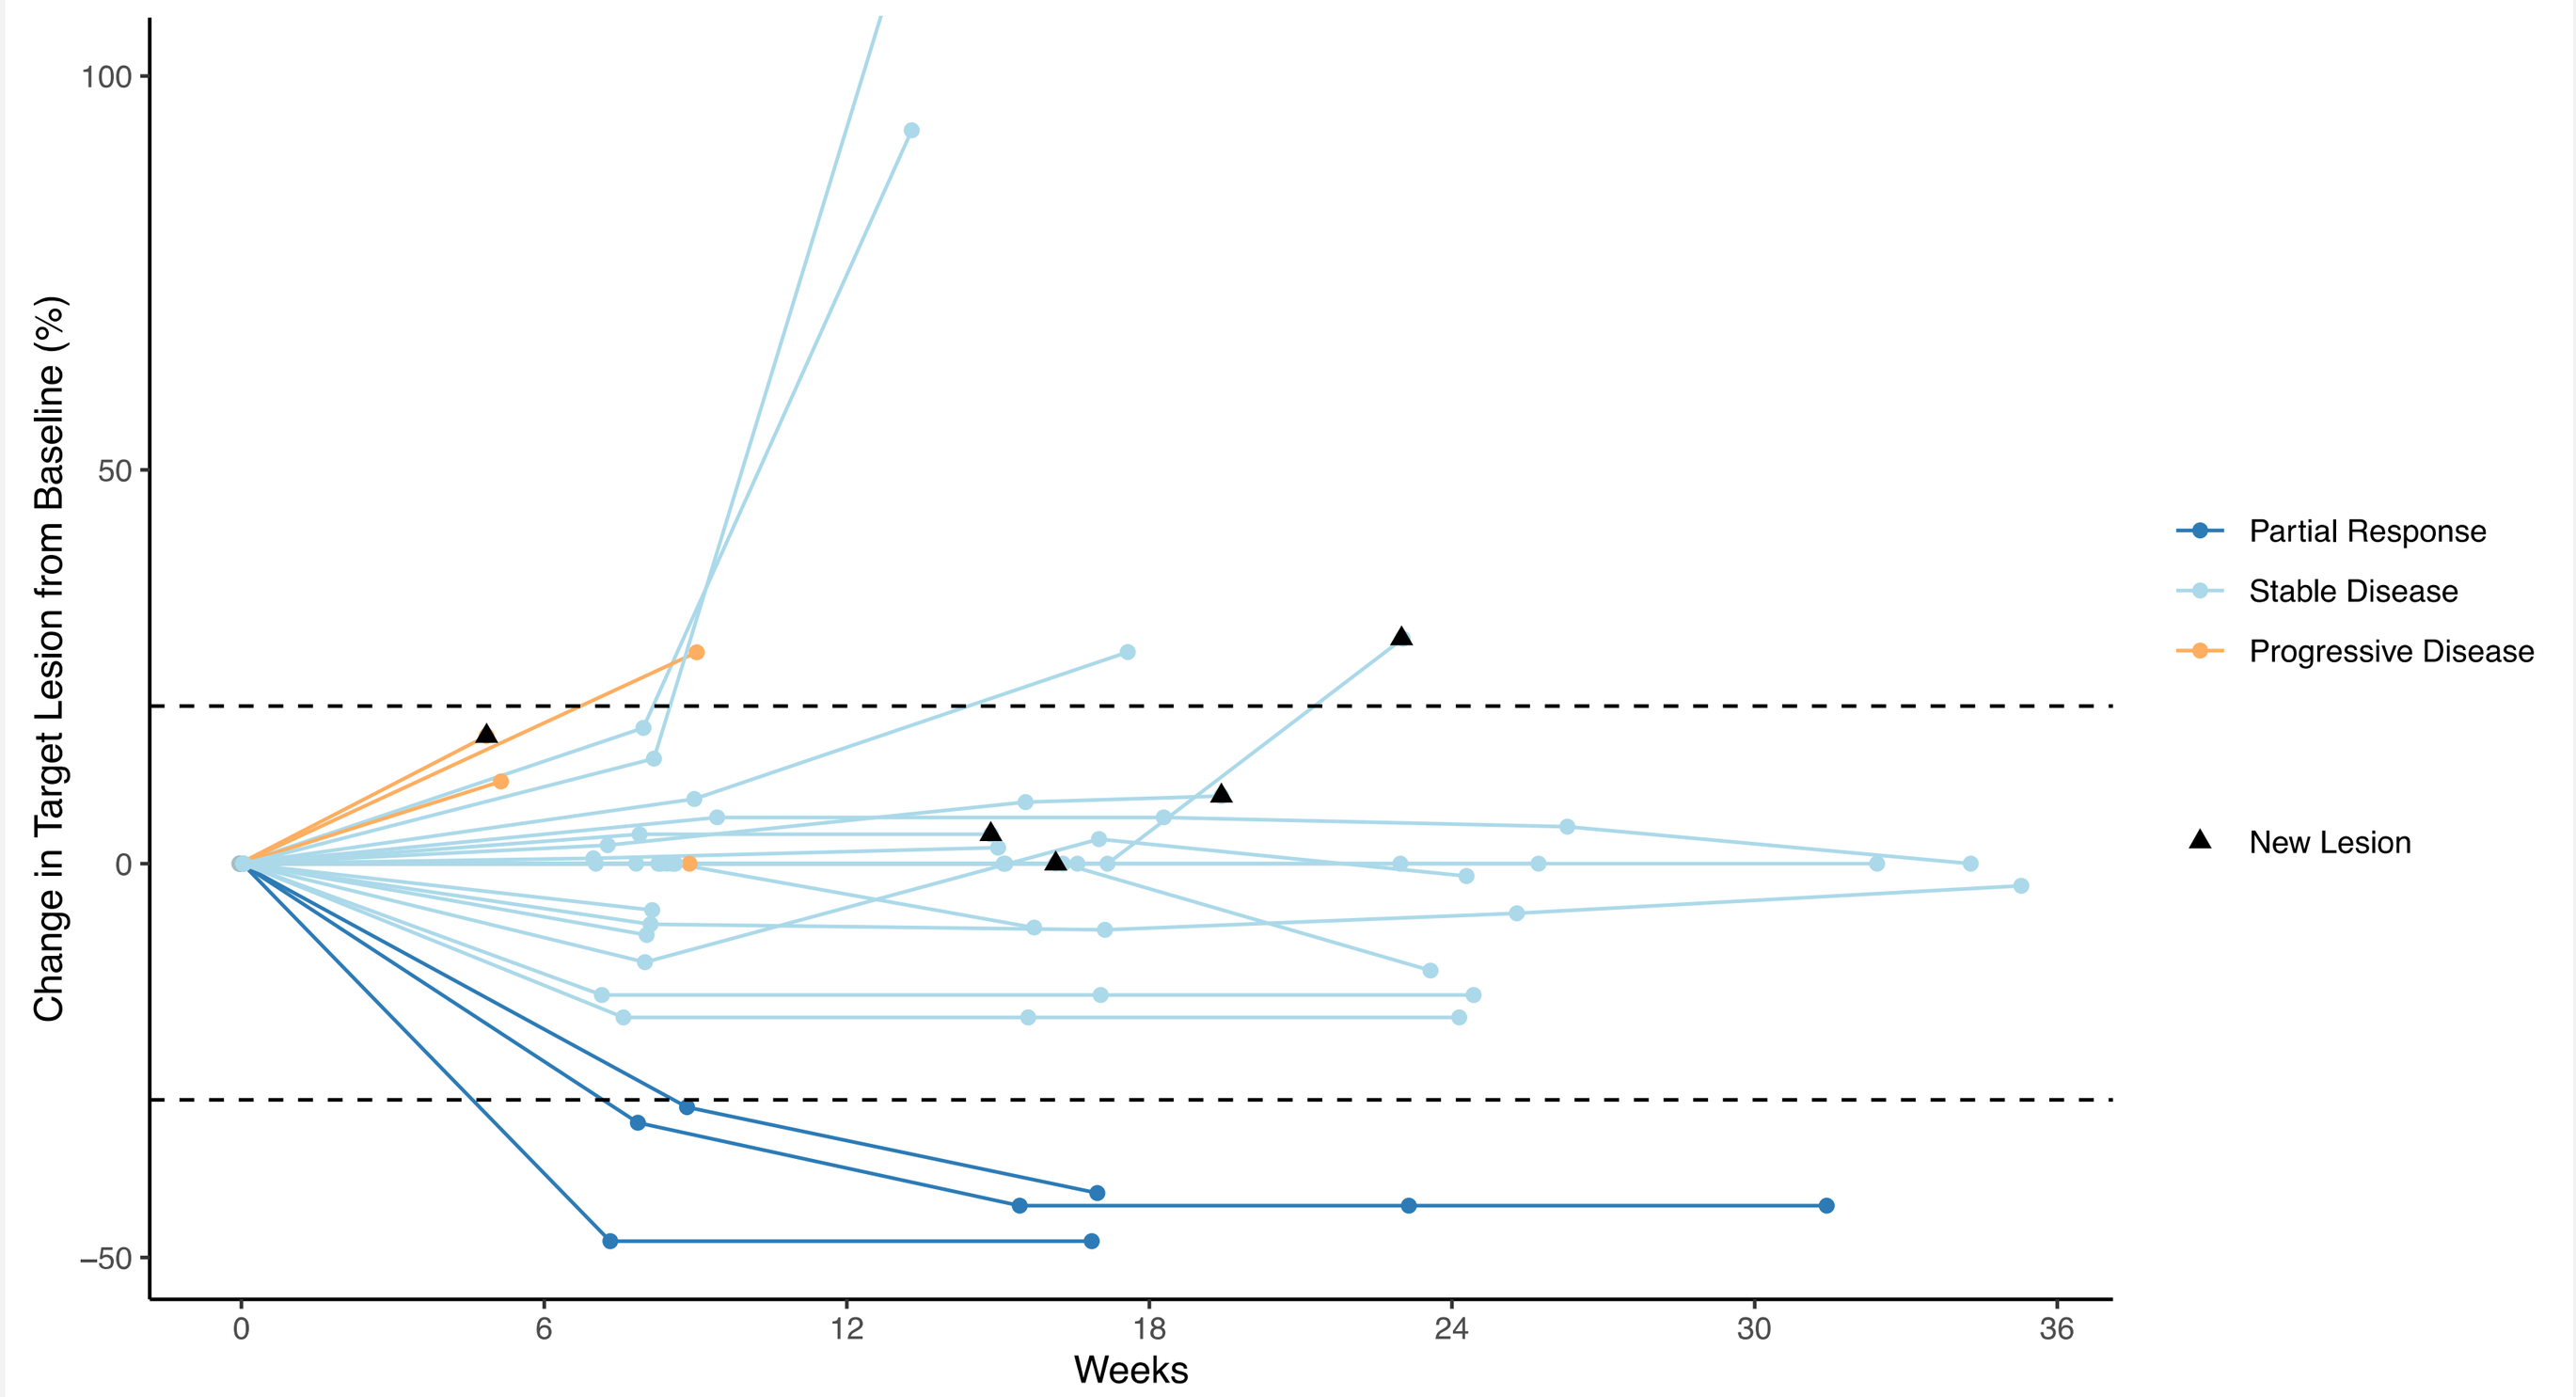
Change in Target Lesion from Baseline (%)
100
50
0
−50
0
6
12
18
24
30
36
Weeks
Partial Response
Stable Disease
Progressive Disease
New Lesion

Table 4. Summary of Efficacy Data

	Gastric Cancer		Biliary Tract Cancer	
	BOLD-100 + FOLFOX	Landmark[2]	BOLD-100 + FOLFOX	Landmark[3]
Progression-Free Survival (PFS), months	5.5 [2.8, 13]	4.6	5.0 [3.1, 8.9]	4.0
Overall Survival (OS), months	15 [5.4, 63]	7.1	7.3 [4.2, 14]	6.2
Overall Response Rate (ORR)	22%	40%	6%	5%
Disease Control Rate (DCR)	89%	69%	83%	33%

2. Mauer AM, Kraut EH, Krauss SA, et al. Phase II trial of oxaliplatin, leucovorin and fluorouracil in patients with advanced carcinoma of the esophagus. *Annals of Oncology*. 2005;16(8):1320-1325. doi:10.1093/annonc/mdi249 3. Lamarca A, Palmer DH, Wasan HS, et al. Second-line FOLFOX chemotherapy versus active symptom control for advanced biliary tract cancer (ABC-06): a phase 3, open-label, randomised, controlled trial. *The Lancet Oncology*. 2021;22(5):690-701. doi:10.1016/S1470-2045(21)00027-9

Gastric Cancer

Figure 3. Median Bayesian Progression-Free Survival (A) and Overall Survival (B) in Metastatic Gastric Cancer

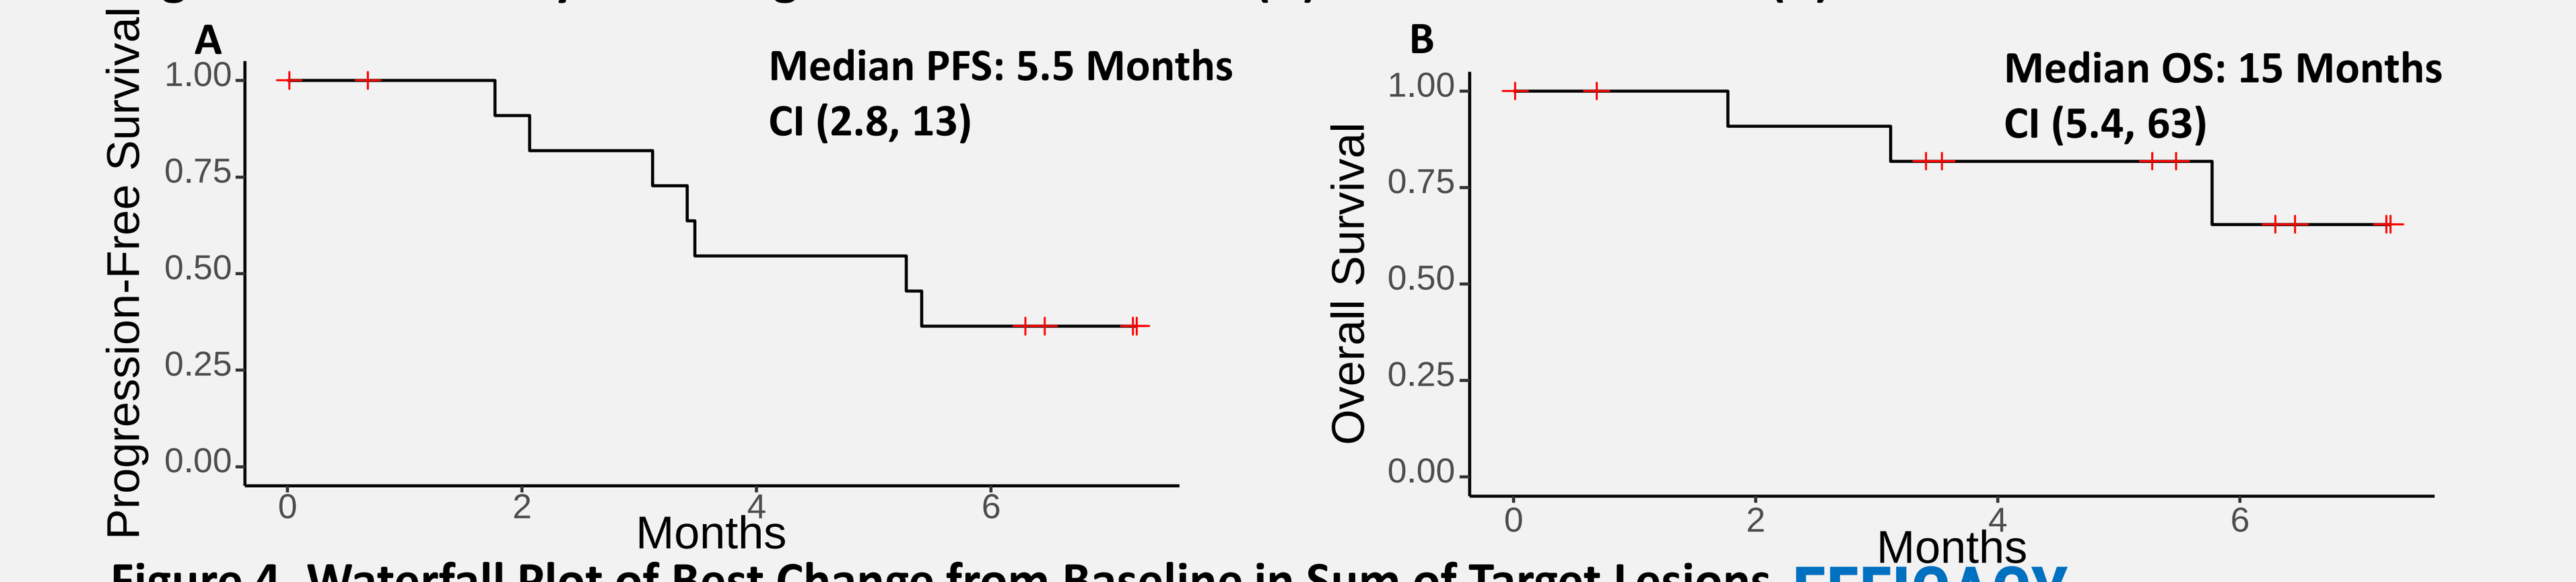
A
Progression-Free Survival
1.00
0.75
0.50
0.25
0.00
0
2
4
6
Months
Median PFS: 5.5 Months
CI (2.8, 13)
B
Overall Survival
1.00
0.75
0.50
0.25
0.00
0
2
4
6
Months
Median OS: 15 Months
CI (5.4, 63)

Figure 4. Waterfall Plot of Best Change from Baseline in Sum of Target Lesions

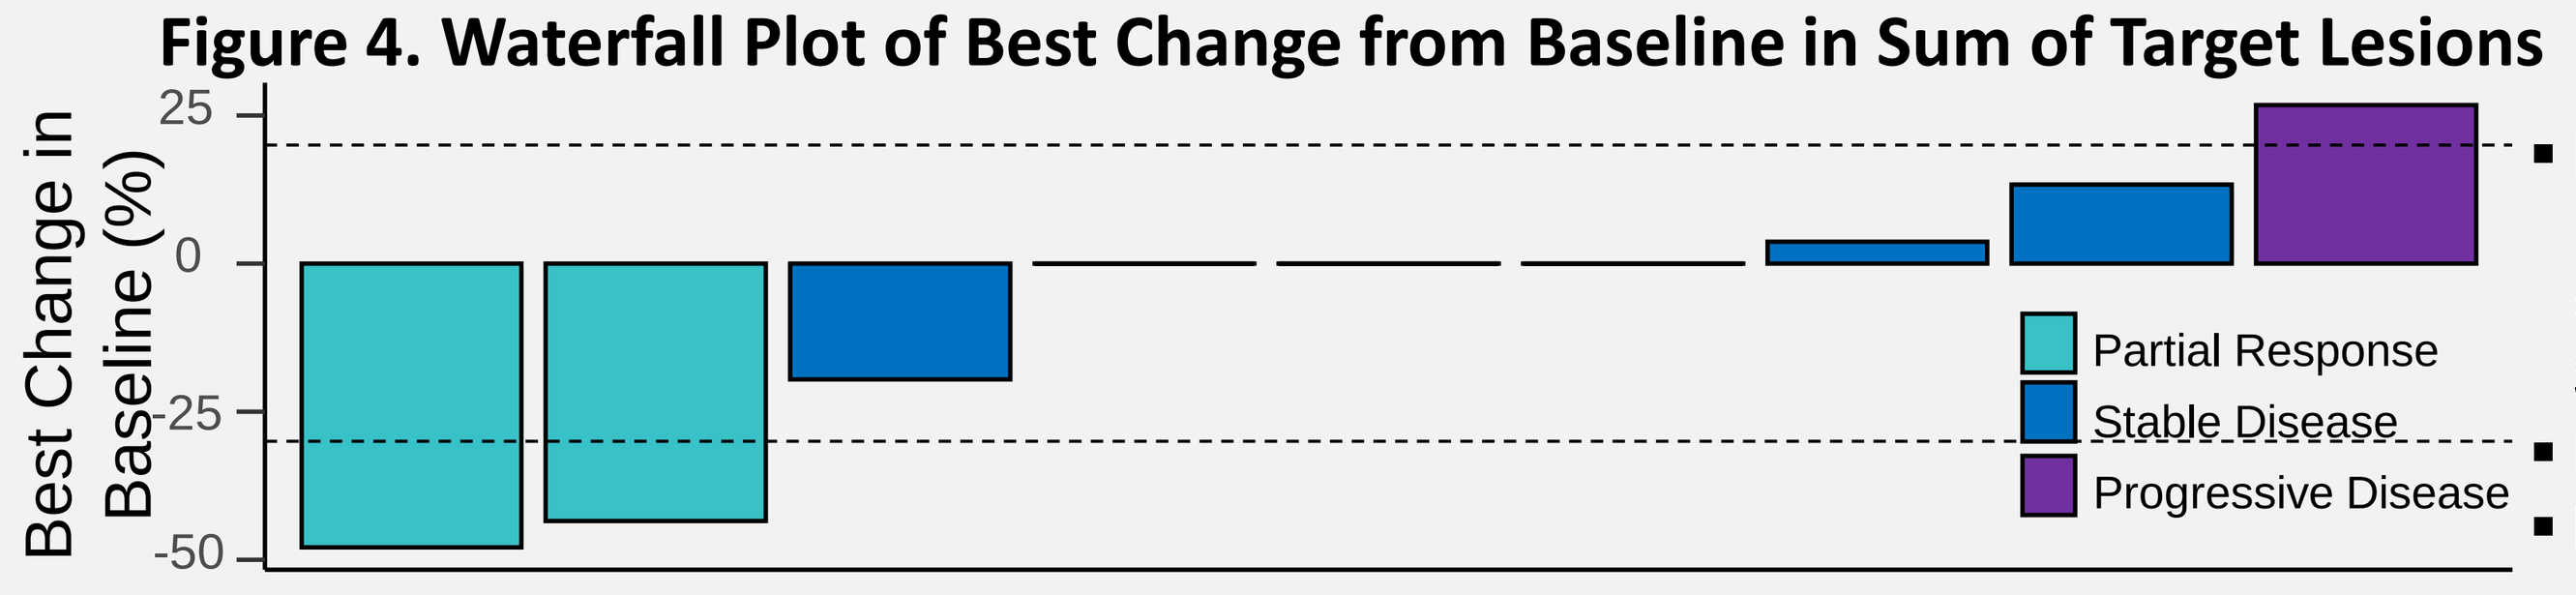
Best Change in Baseline (%)
25
0
-25
-50
Partial Response
Stable Disease
Progressive Disease

2 patients who discontinued the study prior to the first disease assessment are not included in this analysis.

EFFICACY

- Median Bayesian PFS was 5.5 [2.8, 13] months and median Bayesian OS of 15 [5.4, 63] months compares favourably to current SOC.[2]
- PR's were observed in 2/9 patients (ORR: 22%)
- Investigator-assessed DCR was 89%, 2 patients did not have an efficacy assessment.

Biliary Tract Cancer

Figure 5. Median Bayesian Progression-Free Survival (A) and Overall Survival (B) in Metastatic Biliary Tract Cancer

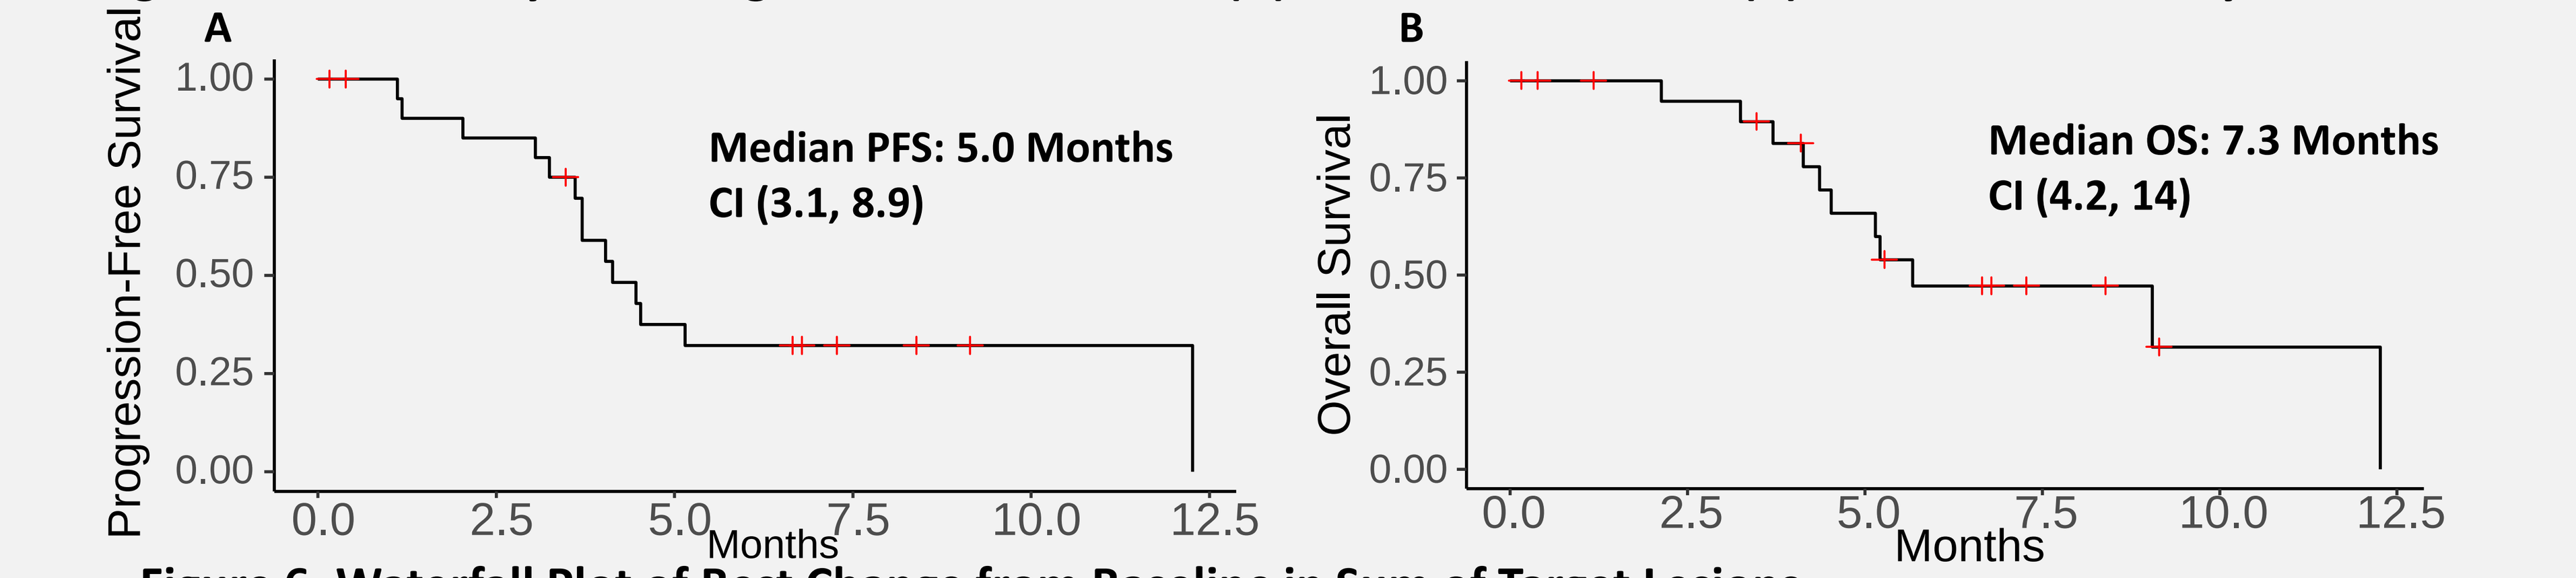
A
Progression-Free Survival
1.00
0.75
0.50
0.25
0.00
0.0
2.5
5.0
7.5
10.0
12.5
Months
Median PFS: 5.0 Months
CI (3.1, 8.9)
B
Overall Survival
1.00
0.75
0.50
0.25
0.00
0.0
2.5
5.0
7.5
10.0
12.5
Months
Median OS: 7.3 Months
CI (4.2, 14)

Figure 6. Waterfall Plot of Best Change from Baseline in Sum of Target Lesions

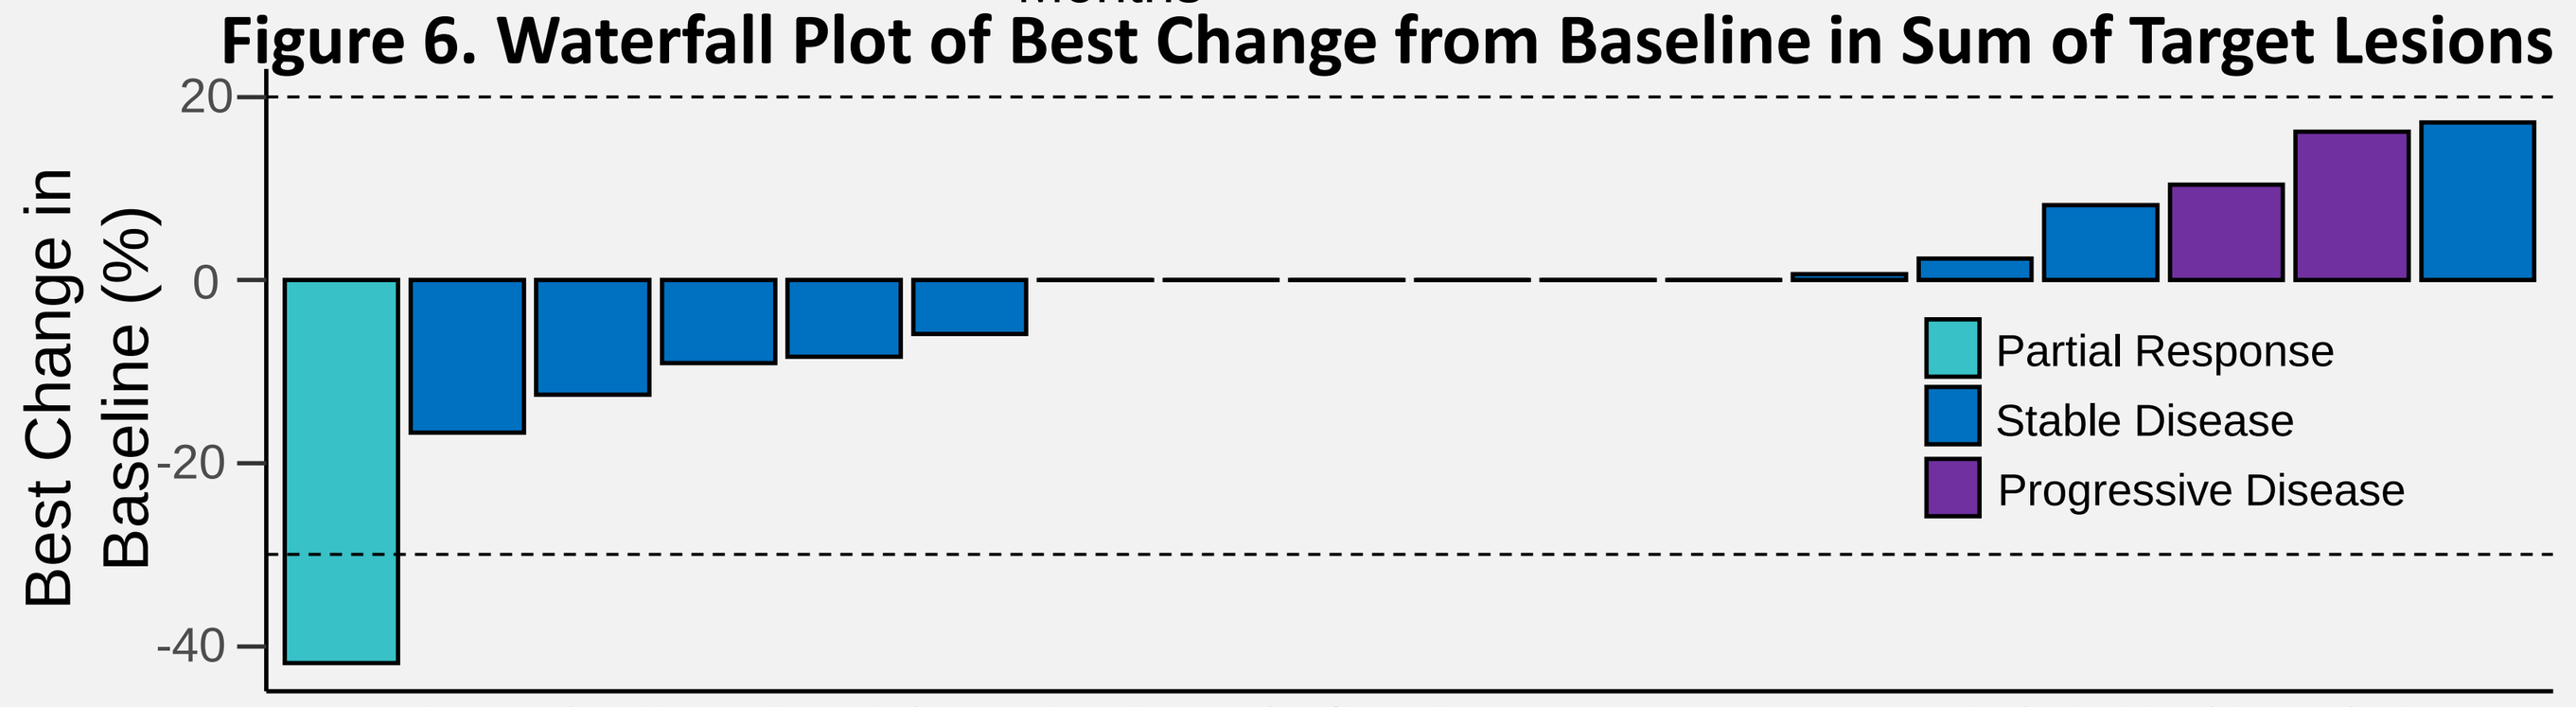
Best Change in Baseline (%)
20
0
-20
-40
Partial Response
Stable Disease
Progressive Disease

4 patients who discontinued the study prior to the first disease assessment are not included in this analysis.

EFFICACY

- Median Bayesian PFS was 5.0 [3.1, 8.9] months and median Bayesian OS of 7.3 [4.2, 14] months compares favourably to current SOC (median PFS 4.0 months; median OS 6.2 months for FOLFOX alone).[3]
- Investigator-assessed DCR was 83%, 4 patients did not have an efficacy assessment.

Conclusions

- The combination of BOLD-100 plus FOLFOX is an active and well tolerated treatment regimen in heavily pre-treated stage IV gastric and biliary tract cancer populations.
- Preliminary PFS, OS, ORR and DCR data in this interim analysis demonstrate significant improvement over currently available therapies for these difficult to treat advanced GI cancers.
- This is interim data with enrollment continuing in the GC arm, and patients in both the GC and BTC arms continue to be treated and followed for efficacy endpoints.

BOLD-100-001 study is sponsored by Bold Therapeutics, Inc.

Contact: jp@bold-therapeutics.com

Presented at American Society of Clinical Oncology (ASCO) in Chicago, IL from June 2-6, 2023

References: 1. Spratlin JL. *JCO*. 2022;40(16_suppl):3031-3031. doi:10.1200/JCO.2022.40.16_suppl.3031
2. Mauer AM. *Annals of Oncology*. 2005;16(8):1320-1325. doi:10.1093/annonc/mdi249
3. Lamarca A. *The Lancet Oncology*. 2021;22(5):690-701. doi:10.1016/S1470-2045(21)00027-9

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.